POSTER #CTNI-20

Addressing Skin Adverse Events During Mirdametinib Treatment in Patients With Neurofibromatosis Type 1-Associated Plexiform Neurofibromas: Guidance From a Multidisciplinary Group of Experts Involved in the ReNeu Trial

Angela Hirbe, MD, PhD[1]; Milan J. Anadkat, MD[1]; Christina Boull, MD[2]; Christopher Moertel, MD[2]; Molly Nickerson, PhD[3]; Michael D. Weber, PharmD[3]; Dusica Babovic-Vuksanovic, MD[4]; and Timothy Gershon, MD, PhD[5]

[1]Washington University School of Medicine, St. Louis, MO, USA; [2]University of Minnesota, Minneapolis, MN, USA; [3]SpringWorks Therapeutics, Inc, Stamford, CT, USA; [4]Mayo Clinic, Rochester, MN, USA; [5]Emory University, Atlanta, GA, USA

Copies of the poster can be obtained through this Quick Response (QR) code for personal use only

INTRODUCTION

Mirdametinib and the ReNeu trial

- Plexiform neurofibromas (PN) are nonmalignant tumors reported in 30% to 50% of patients with neurofibromatosis type 1 (NF1) that often cause significant morbidity[1-4]
- Mirdametinib is an investigational, oral, allosteric, highly selective, potent, central nervous system-penetrant, small-molecule inhibitor of mitogen-activated protein kinase kinase 1 and 2 (MEK1/2)[5-7]
- In the multicenter, pivotal, phase 2b ReNeu trial (NCT03962543), mirdametinib demonstrated deep and durable PN volume reductions, improvement in pain and health-related quality of life by patient and parent proxy-reports, and a manageable safety profile in adults (≥18 years of age; n=58) and children (2 to 17 years of age; n=56) with NF1-PN
- Patients received mirdametinib 2 mg/m^2 twice a day (BID; max 4 mg BID) for 21 days of each 28-day cycle (total 24 cycles); no fasting requirement[8]
- Skin treatment-related adverse events (TRAEs) were commonly reported in ReNeu (**Table 1**)
- Skin adverse events (AEs) are among the most common AEs reported with MEK inhibitors[9-11]
 - Commonly reported MEK inhibitor-induced skin AEs include acneiform rash (particularly in postpubescent patients) and eczematous dermatitis (particularly in prepubescent patients)[12]

Table 1. Skin TRAEs Commonly Reported in the ReNeu Trial

	PATIENTS (N=114), %
≥1 skin TRAE	
Adults	93
Children	82
Dermatitis acneiform[a]	
Adults	78
Children	43
Skin TRAE leading to dose modification, % adults/children	
Treatment interruption	0/2
Dose reduction	10/4
Permanent discontinuation	14/7

[a]Most common skin TRAE; most events began during cycle 1 or 2. Skin TRAEs included the following (in order of decreasing incidence in the trial population): dermatitis acneiform, dry skin, alopecia, rash, eczema, pruritus, hair color changes, urticaria, hair texture abnormal, rash maculopapular, acne, pain of skin, nail disorder, onychomadesis, photosensitivity reaction, seborrheic dermatitis, skin fissures, dermatitis, erythema, exfoliative rash, follicular eczema, hyperkeratosis, nail dystrophy, abnormal nail growth, onychalgia, palmar-plantar erythrodysesthesia syndrome, papule, rash erythematous, rash papular, rash pruritic, skin burning sensation, skin exfoliation, skin hypopigmentation, skin wrinkling, and trichorrhexis. TRAE, treatment-related adverse event.

Mechanism of MEK inhibitor-induced skin adverse events

- Understanding the mechanism of MEK inhibitor-induced skin AEs can help us understand and create better ways to mitigate MEK inhibitor-induced rashes experienced in clinical practice
- MEK inhibitors can disrupt the skin barrier and impair antibacterial defense, leading to papulopustular rash, pruritus, xerosis, and folliculitis (**Figure 1A-1C** [non-ReNeu patients])[13]
- MEK inhibitors may create a permissive environment for *Cutibacterium acnes* within the hair follicle, driving an innate immune response mediated by keratinocyte-derived nuclear factor-κB, interleukin (IL)-36γ, and IL-8[14,15]
- These proinflammatory mediators recruit neutrophils, resulting in folliculocentric pus and characteristic MEK inhibitor-induced acneiform rash (**Figure 1D** and **1E** [non-ReNeu patients])[14,15]

Figure 1. Typical MEK Inhibitor-Induced Skin AEs From Non-ReNeu Pre- and Postpubescent Patients

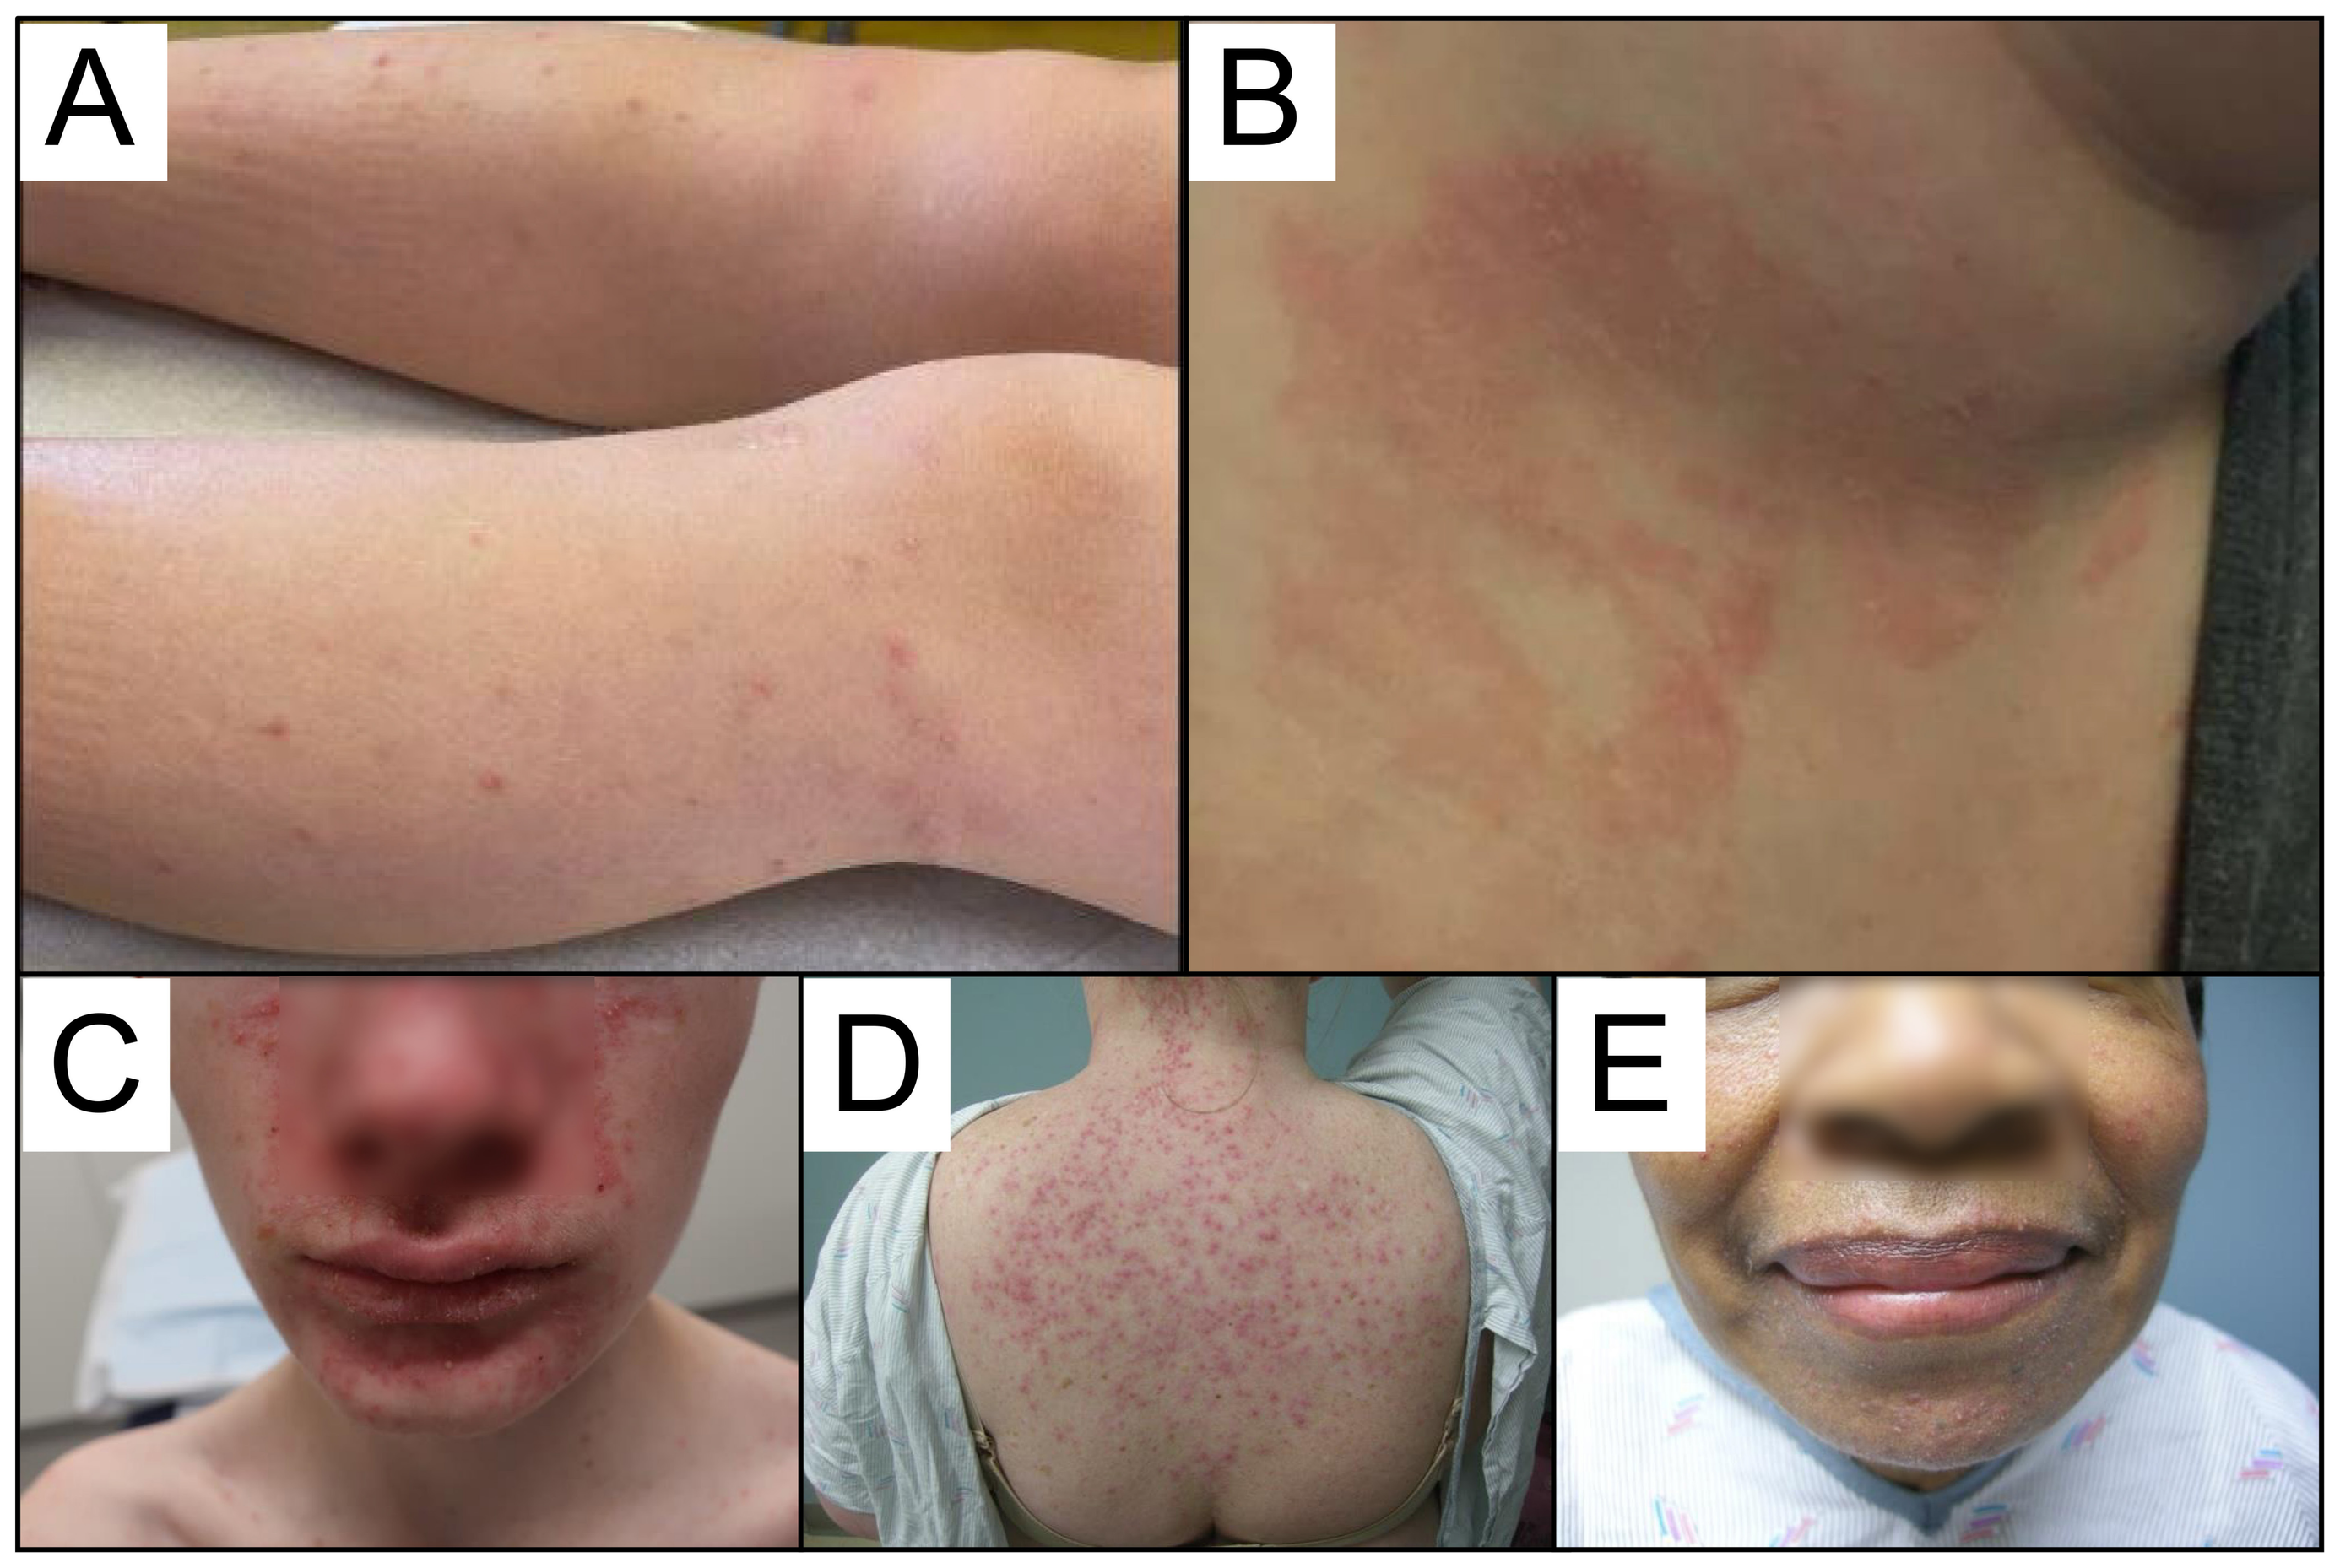

A
B
C
D
E

A, papular variant of dermatitis on prepubescent patient; B, plaque of dermatitis on the anterior neck and chest of prepubescent patient; C, acneiform rash on young postpubescent patient; D, grade 2 rash on adult patient; E, rash on adult patient. Reprinted with permission from Milan J. Anadkat and Christina Boull.

OBJECTIVES

- The goal is to share skincare management recommendations for mirdametinib developed by the ReNeu Scientific Steering Committee
- These recommendations are being shared to highlight potential proactive and reactive supportive care to consider while managing patients on MEK inhibitors
- These recommendations may help to improve the medical management of skin TRAEs and to reduce their incidence and severity, while decreasing dose modifications to ultimately keep patients on mirdametinib

METHODS

- In ReNeu, physical examinations of the skin were conducted throughout the study, and skin TRAEs were graded using the National Cancer Institute Common Terminology Criteria for Adverse Events, version 5.0[16]
- The ReNeu Scientific Steering Committee (ReNeu investigators and expert dermatologists) retrospectively reviewed skincare management at one high-enrolling ReNeu trial site (n=12; 19-68 years of age) where no mirdametinib dose modifications were needed due to skin TRAEs to develop skincare recommendations for patients receiving mirdametinib within the ReNeu clinical trial
- These skincare management recommendations were incorporated into the ReNeu trial recommendations and recently updated in the present recommendations
 - These recommendations were not part of the study protocol

Figure 2. Skincare Management Recommendations for ReNeu Trial Patients

Skincare Recommendations for All Patients

- Patients are advised to optimize general skincare to prevent and limit dermatologic reactions including dry skin, which can also be associated with mirdametinib
- Soap usage should be limited to essential areas (eg, face, axillae, groin, and between toes)
- Dry skin may predispose patient to increased risk of injuries and fissures resulting in infection[9,18]
- All patients are counseled to adopt hygienic skincare practices, including bathing daily and using mild cleansers and hypoallergenic skin moisturizers (cream or ointment-based emollient; minimum BID) to prevent dry skin

PARTICIPANT GROUP	PREPUBESCENT PATIENTS		POSTPUBESCENT PATIENTS
ALL PATIENTS UPON MIRDAMETINIB INITIATION	**Directions for bleach baths should be provided to patients and can begin with mirdametinib treatment or in reaction to the development of rash** ▪ Add ¼ to ½ cup of regular strength bleach to a full bathtub (~1 teaspoon per gallon) and soak for 5-10 minutes 3-4× per week[9] ▪ Follow immediately with high-quality hypoallergenic skin moisturizer ▪ Patients are instructed to strictly follow guidance for bleach baths and avoid contact of nondiluted bleach with eyes, which can cause irreversible eye damage and skin burns[19]		**Prophylactic treatment with topical clindamycin and an oral tetracycline at an anti-inflammatory dose is recommended for prevention and reduced severity of acneiform rash[22,23]** ▪ Topical clindamycin lotion (1.0% BID) for face ▪ Begin treatment with a tetracycline (eg, doxycycline or minocycline; 50 mg/day for 3 months)
MILD RASH REACTIVE TREATMENT	DERMATITIS **Generally, treatments recommended for mild-to-severe dermatitis are hydrocortisone (2.5% BID, weaker) and triamcinolone (0.1% BID, stronger) ointment or cream formulations** Specifically, ▪ Hydrocortisone cream (2.5% BID) for face and skinfold areas ▪ Triamcinolone ointment (0.1% BID) for trunk and extremities ▪ If the condition does not resolve within 2 weeks, consider consulting a dermatologist	ACNEIFORM RASH **Acneiform rash occurred infrequently in prepubescent patients treated with mirdametinib; however, acneiform rash unrelated to treatment may develop in pubescent patients, requiring a differential diagnosis to appropriately attribute symptoms to puberty or treatment-emergent rash[20]** Treatments for mild acneiform rash include: ▪ Topical clindamycin lotion (1.0% BID) until rash is clear and as needed thereafter ▪ Hydrocortisone cream (2.5% BID) can be added	ACNEIFORM RASH If acneiform rash still develops ***and*** is bothersome, add ▪ Hydrocortisone cream (2.5% BID) **OR** ▪ Triamcinolone ointment (0.1% BID)
MODERATE TO SEVERE RASH REACTIVE TREATMENT		ACNEIFORM RASH The following treatments options are suggested for moderate to severe acneiform rash: ▪ Cephalexin (20 mg/kg/day divided BID; max dose 500 mg) up to 6 weeks **OR** ▪ Amoxicillin (25 mg/kg/day divided BID; max dose 875 mg) up to 6 weeks **OR** ▪ Fluconazole (20 mg/kg/day; max dose 100 mg) for 5 days and then 1× per week for 3 months[21] ▪ Referral to a dermatologist should be made at initiation of cephalexin or amoxicillin treatment	
REFERRAL TO DERMATOLOGY	▪ Dermatitis uncontrolled with recommended supportive care	▪ For moderate/severe acneiform rash concurrent with initiation of cephalexin/amoxicillin, or suspected infection	▪ If acneiform rash continues to worsen or if an infection is suspected, refer to dermatologist - Stronger topical steroids or oral retinoids may be added ▪ Skin that is fissured may be susceptible to superinfection, which is associated with pain instead of itching[24] ▪ Patients should consult a dermatologist for worsening infectious or noninfectious rash (eg, erythematous rash)
ADDITIONAL PRECAUTIONS	▪ If interventions for mild to severe rash are unsuccessful, all patients with uncontrolled dermatitis, worsening acneiform rash, or infection should be referred to a dermatologist ▪ Avoid agents with the potential to dry skin, such as benzoyl peroxide, salicylic acid, acne skin washes, scrubs, exfoliants, anti-aging creams, alcohol (cleansers, wipes) or other agents that can dry skin ▪ Avoid topical retinoids		

AE, adverse event; BID, twice a day.

GUIDANCE

- Skin AEs associated with mirdametinib may vary by pubertal status; therefore, skincare management recommendations are different for prepubertal and postpubertal patients (**Figure 2**)[12,17]
- Healthcare providers should obtain a detailed history of skin disorders before initiating treatment to identify patients with a higher risk of developing skin AEs (eg, history of eczema, ichthyosis, or other conditions that cause dry skin)
 - Preventive therapy may be indicated for these patients
- Consultation with a dermatologist is recommended for uncontrolled dermatitis or worsening acneiform rash and infection
- For mild-to-severe dermatitis, the important factor in topical steroid choice is strength (hydrocortisone 2.5% is considered to be least potent among steroids and triamcinolone 0.1% is considered to be of medium potency among steroids), whereas the choice between vehicle (cream or ointment) is driven by patient preference

DISCUSSION

- This guidance on the management of common skin TRAEs in the ReNeu clinical trial was implemented late in the trial, and outcomes measuring the success of this guidance are not available
- Acneiform rash is common in postpubescent patients treated with mirdametinib; most cases arise within the first 2 cycles of treatment and may be mitigated with proactive management[17]
- The prophylactic use of systemic antibiotics for MEK inhibitor-induced moderate to severe acneiform rash has not been prospectively validated

CONCLUSIONS

- The ReNeu Scientific Steering Committee, in collaboration with expert dermatologists, created these skincare recommendations to proactively and reactively manage potential skin TRAEs for patients treated with mirdametinib in the ReNeu trial
- Optimization of skin TRAE management during MEK inhibitor treatment is hypothesized to help reduce AE incidence and severity and decrease dose modifications, while improving the patients' journey through treatment
- This experience with the ReNeu trial highlights an unmet medical need for management of MEK inhibitor-associated skin AEs and better guidance in clinical practice and trials, which could improve patient experience with MEK inhibitor treatment

REFERENCES

1. Miller DT, et al. *Pediatrics*. 2019;143(5):e20190660. **2.** Prada CE, et al. *J Pediatr*. 2012;160(3):461-7. **3.** Fisher MJ, et al. *Neuro Oncol*. 2022;24(11):1827-44. **4.** Gutmann DH, et al. *Nat Rev Dis Primers*. 2017;3:17004. **5.** de Gooijer MC, et al. *Int J Cancer*. 2018;142(2):381-91. **6.** Jousma E, et al. *Pediatr Blood Cancer*. 2015;62(10):1709-16. **7.** LoRusso PM, et al. *Clin Cancer Res*. 2010;16(6):1924-37. **8.** Moertel CL, et al. *J Clin Oncol*. 2024;42(16_suppl):3016. **9.** Klesse LJ, et al. *Oncologist*. 2020;25(7):e1109-e16. **10.** Russo I, et al. *Scientifica (Cairo)*. 2018;2018:5036213. **11.** Welsh SJ, Corrie PG. *Ther Adv Med Oncol*. 2015;7(2):122-36. **12.** de Blank PMK, et al. *Neuro Oncol*. 2022;24(11):1845-56. **13.** Jiménez-Andrade Y, et al. *Hematol Oncol Clin North Am*. 2024;38(5):1011-25. **14.** Billi AC, et al. *J Clin Invest*. 2020;130(3):1090-2. **15.** Satoh TK, et al. *J Clin Invest*. 2020;130(3):1417-30. **16.** US Department of Health And Human Services. 2017. https://ctep.cancer.gov/protocoldevelopment/electronic_applications/docs/CTCAE_v5_Quick_Reference_5x7.pdf. **17.** Yuan C, Wang B. *J Cosmet Dermatol*. 2023;22(8):2150-7. **18.** Owczarek W, et al. *Postepy Dermatol Alergol*. 2017;34(5):418-28. **19.** Chu DK, et al. *Ann Allergy Asthma Immunol*. 2024;132(3):274-312. **20.** Nair PA, et al. *StatPearls [Internet]*. Treasure Island, FL: StatPearls Publishing; 2024. **21.** Steele C, et al. *JAMA Dermatol*. 2020;156(6):706-8. **22.** Garner SE, et al. *Cochrane Database Syst Rev*. 2012;2012(8):CD002086. **23.** Gorji M, et al. *Asia Pac J Clin Oncol*. 2022;18(6):526-39. **24.** Specht S, Persaud Y. *StatPearls [Internet]*. Treasure Island, FL: StatPearls Publishing; 2024.

ACKNOWLEDGMENTS

The study was sponsored by SpringWorks Therapeutics, Inc. Medical writing and editing assistance were provided by Cathy R. Winter, PhD, and Stephen Bublitz, ELS, of MedVal Scientific Information Services, LLC (Princeton, NJ) and were funded by SpringWorks Therapeutics, Inc.

DISCLOSURES

AH: Consultancy/advisory role: Alexion Pharmaceuticals, AstraZeneca, Intellisphere, LLC, and SpringWorks Therapeutics, Inc.; patents, royalties, or other intellectual property: Boehringer Ingelheim RCV GmBH & Co KG- Licensing- T-019044 Development of a Preclinical NF1-MPNST Platform Suitable for Precision Oncology Drug Discovery and Evaluation, paid through institution; Deutsches Krebsforschungszentrum-licensing agreement for PDX cell lines, paid through institution; travel expenses: Alexion Pharmaceuticals and SpringWorks Therapeutics, Inc. **MJA:** Personal fees as a past consultant: AbbVie, Adgero, Amgen, Aspire Bariatrics, AstraZeneca, Biogen, Bristol Myers Squibb, Eli Lilly, ImClone, Innovaderm Research, OnQuality Pharmaceuticals, Therakos, UCB Pharma, and Veloce; current or recent advisor and/or consultant: BioLinq, Boehringer-Ingelheim, Novocure, OnQuality Pharmaceuticals, Protagonist, and SpringWorks Therapeutics, Inc.; Principal Investigator: AbbVie, AnaptysBio, Biogen, Boehringer-Ingelheim, Chemocentryx, Eli Lilly, Hana Biosciences, InCyte, InflaRx, Lutris, Moberg, MoonLake, Novartis, OnQuality Pharmaceuticals, Phoenecis, Regeneron, UCB Pharma, Veloce, Xbiotech, and Xoma. **CB:** Consultant: SpringWorks Therapeutics, Inc.; royalties: Springer Nature Group and Wolters Kluwer. **CM:** Employment: OX2 Therapeutics; leadership role: OX2 Therapeutics; equity interest: OX2 Therapeutics; consultancy/advisory role: Alexion Pharmaceuticals; patents, royalties, or other intellectual property: OX2 Therapeutics; travel expenses: Alexion Pharmaceuticals. **MN and MDW:** Employment: SpringWorks Therapeutics, Inc. **DB-V:** Employment: Mayo Clinic; consultancy/advisory role: Alexion Pharmaceuticals; research funding: Alexion Pharmaceuticals, Recursion, and SpringWorks Therapeutics, Inc. **TG:** Research funding: SpringWorks Therapeutics, Inc.

Presented at the 29th Annual Meeting of the Society for NeuroOncology, November 21-24, 2024, Houston, TX, USA